P.1.h.016

Measuring sociability of mice using a novel 3-chamber set-up and algorithm of an automated animal behaviour analysis system

Krisztina Sz. Kordás[1], Herman Eldering[2], Ronald Bulthuis[2], Viktor Román[1]

[1]*Laboratory of Neurodevelopmental Biology, Gedeon Richter Plc., Budapest, Hungary*

[2]*Metris b.v., Hoofddorp, the Netherlands*

Background:

Autism spectrum disorder is characterized by impaired sociability as one of the major symptoms in humans. The 3-chamber sociability test is a widely accepted animal behavioural assay which can be used to examine social behaviour of rodents [1,2].

Aim of the study:

Here we designed and validated a new algorithm to measure social preference of mice in an automated behavioural analysis system (LABORAS™, Metris b.v., the Netherlands) [3]. The system allows parallel automated observations therefore, time and labour consuming observational work can be replaced. Validation involved the investigation of three different mouse strains with various levels of social behaviour and the effects of drugs (3,4-methylenedioxy-methamphetamine (MDMA) and phencyclidine (PCP)). Furthermore, it was also investigated how aggression contributes to social behaviour in this assay.

Methods:

As a validation, we compared the results of the system with the scores of two independent blinded observers using parallel recorded videos. Time spent in chambers and in contact zones around the social/non-social cylinders were determined (Fig. 1). During validation we optimized contact zone radius to find the one most matching with observers' scores. Optimized contact zone radius of the set-up was chosen to be 90 mm (Fig. 2). As a target mouse DBA/2 strain was used and placed into the cylinders, except when DBA strain was measured wherein NMRI mice were used. Male mice weighing 25-40 g were obtained from the following breeders: NMRI from ToxiCoop, Hungary; C57Bl6/J and DBA/2 from Envigo (formerly Harlan), Germany/UK, and CD-1 from Charles River, Germany. Drugs and vehicle (phys. saline) were administered intraperitoneally in a volume of 10 ml/kg. Statistical analysis was performed with GraphPad Instat or GraphPad Prism (GraphPad, San Diego, USA).

Validation of the algorithm *in order to find the best matching parameters with observers' scores*

Fig. 1

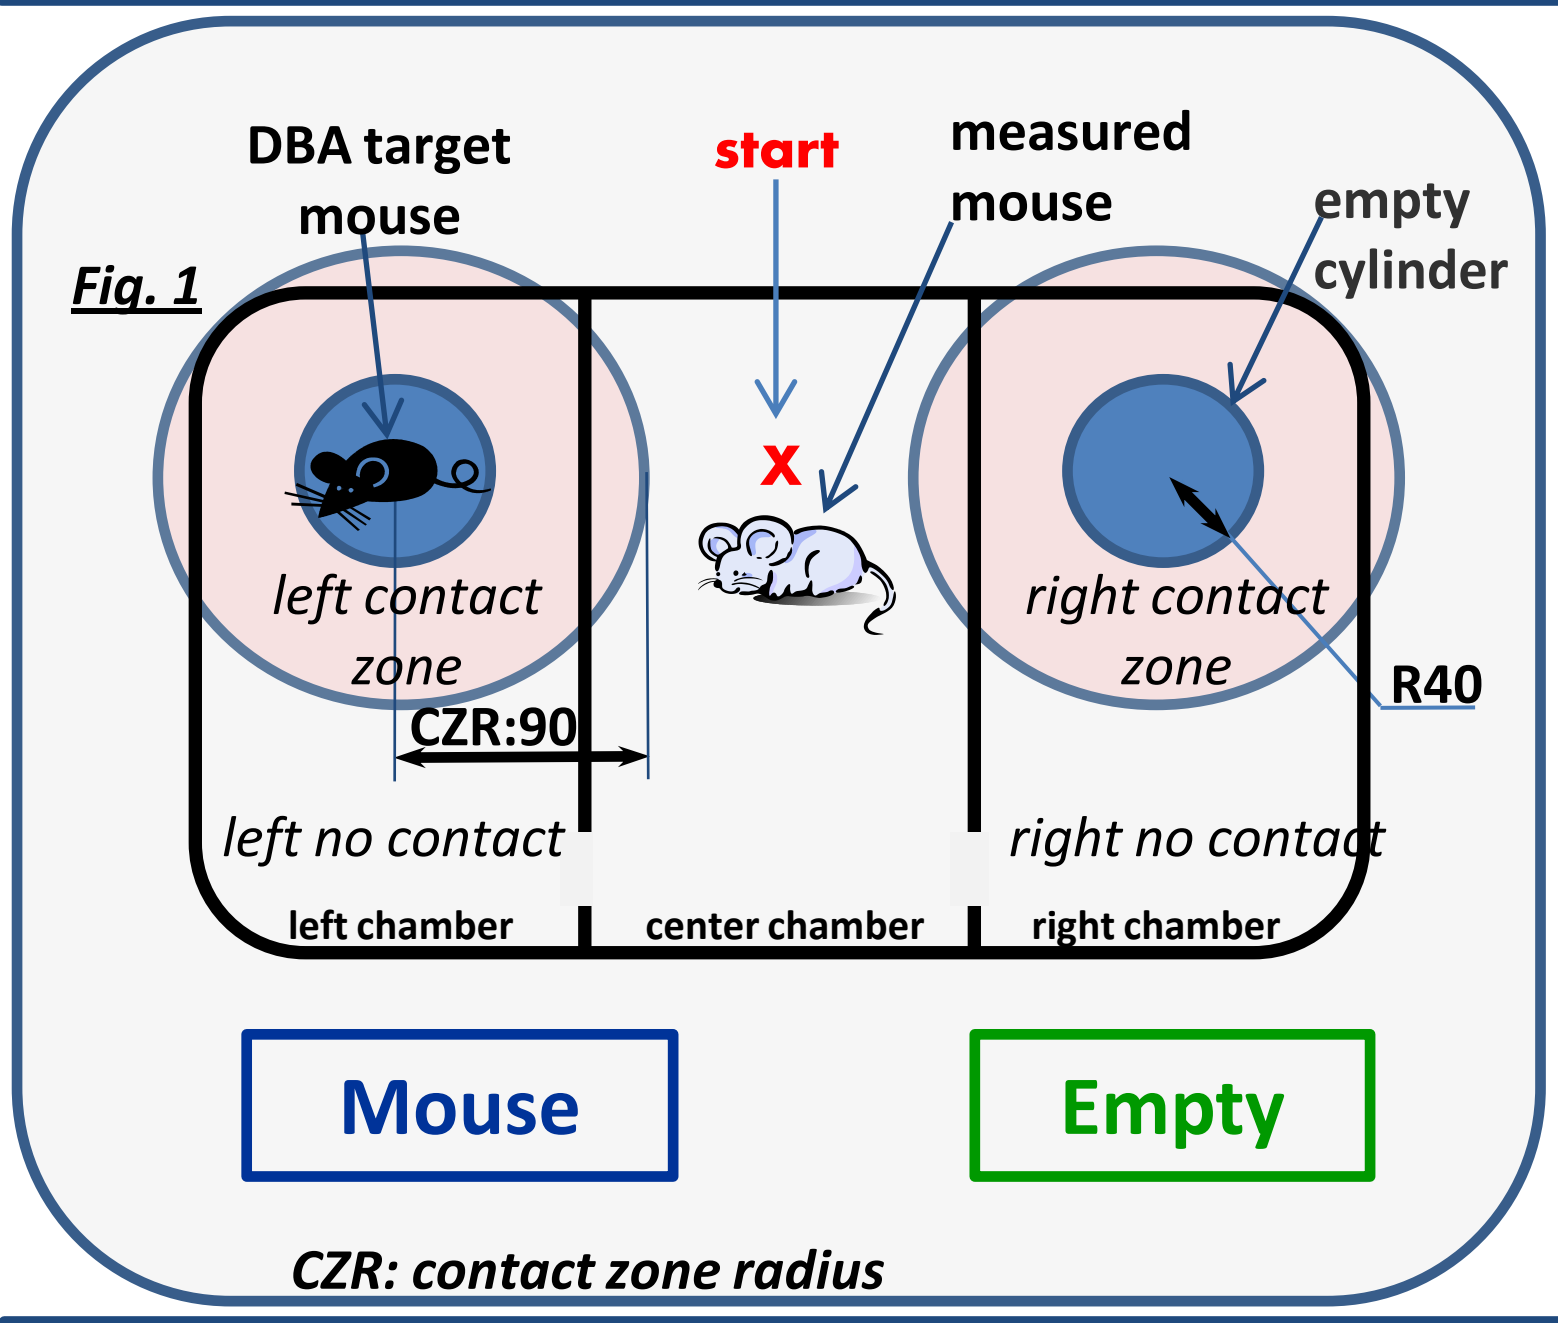

DBA target mouse
start
measured mouse
empty cylinder
X
left contact zone
right contact zone
R40
CZR:90
left no contact
right no contact
left chamber
center chamber
right chamber
Mouse
Empty

CZR: contact zone radius

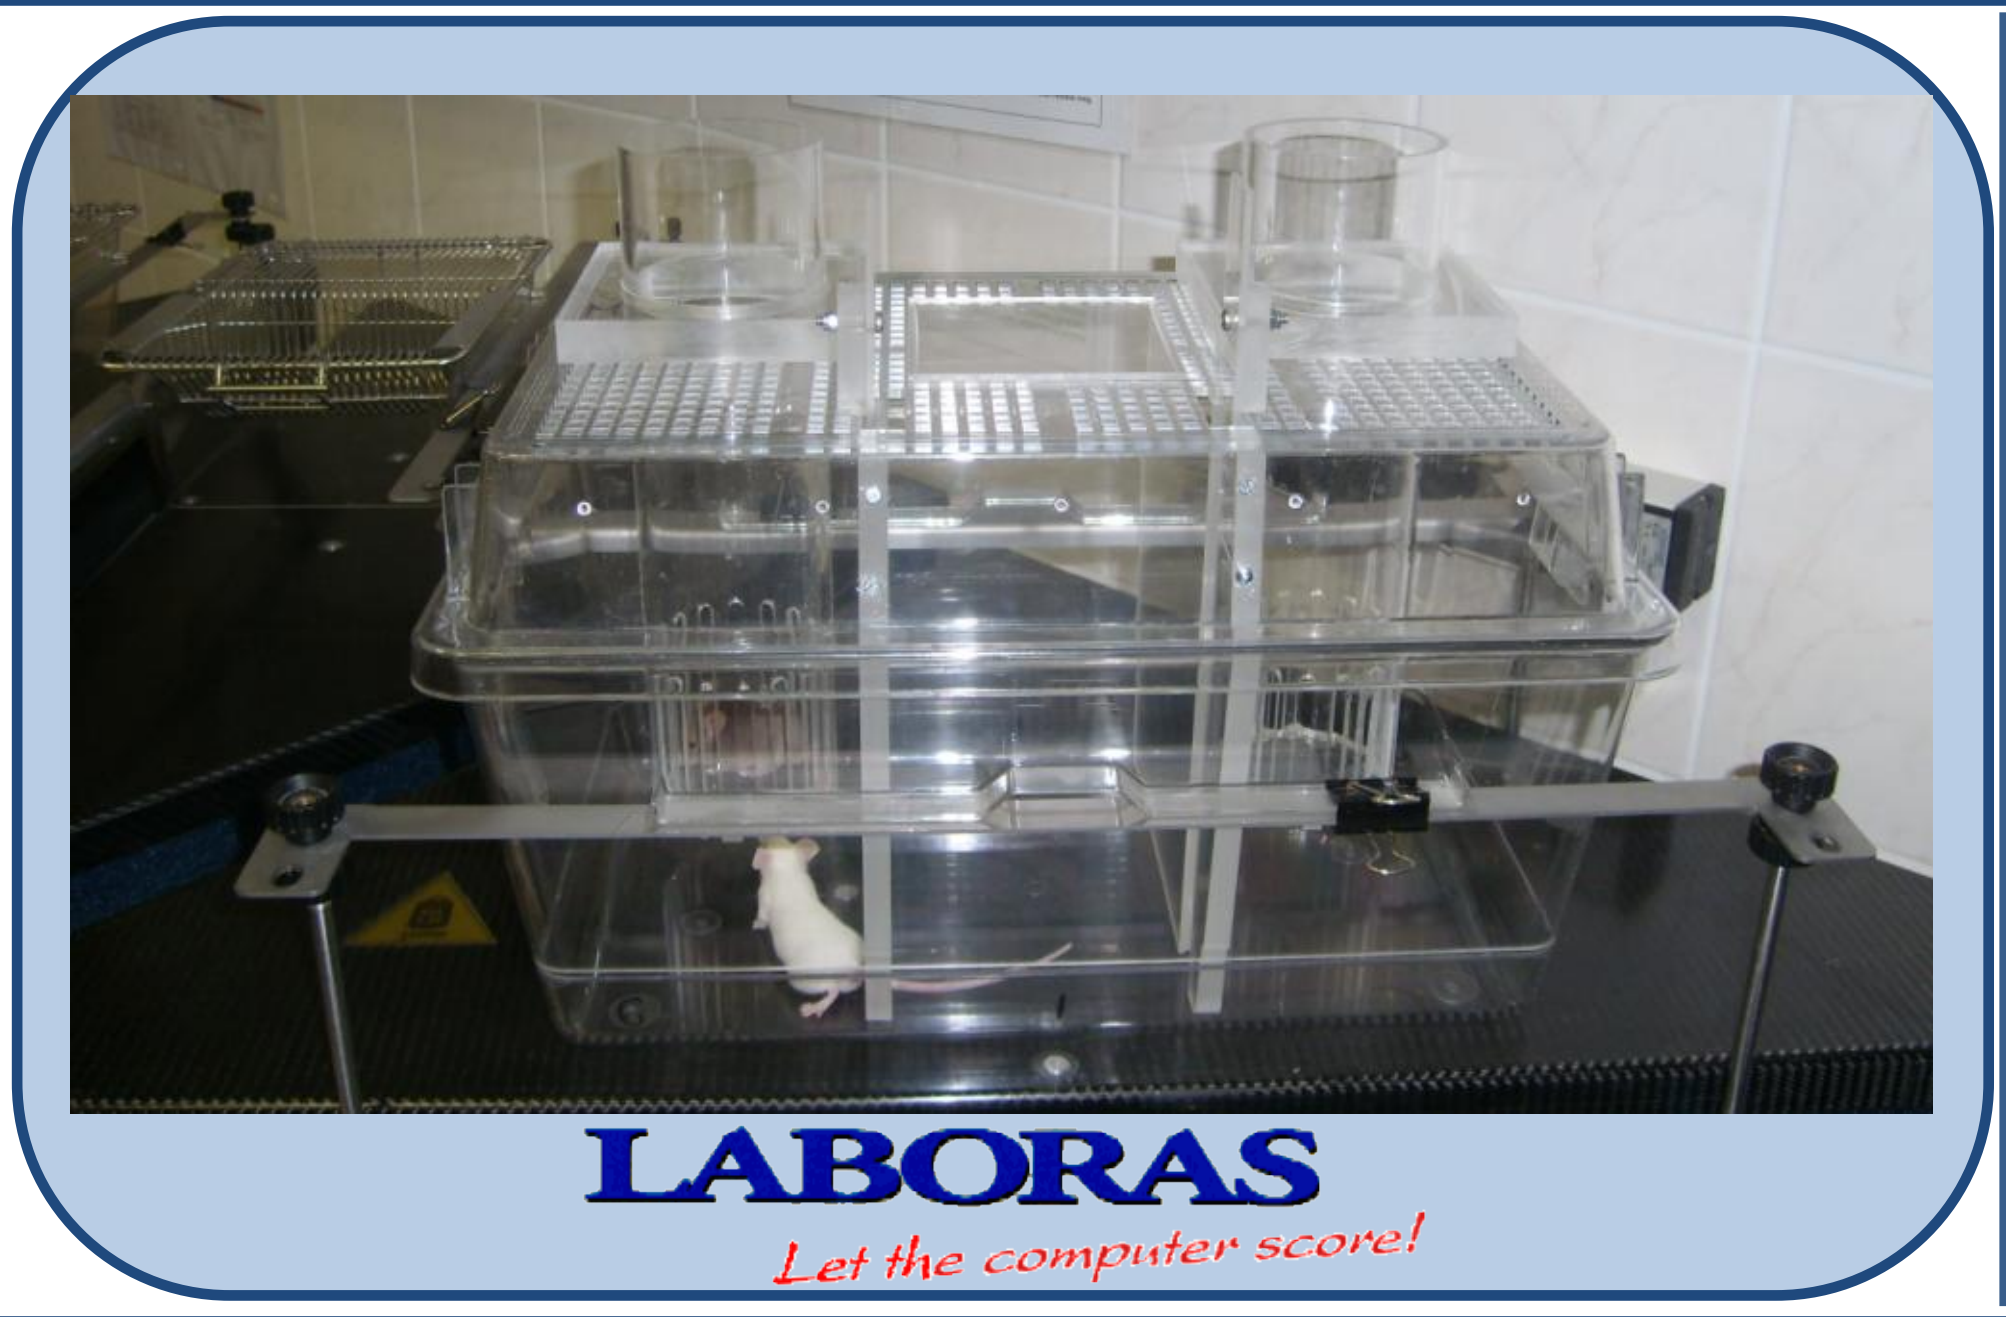

LABORAS
Let the computer score!

Fig. 2

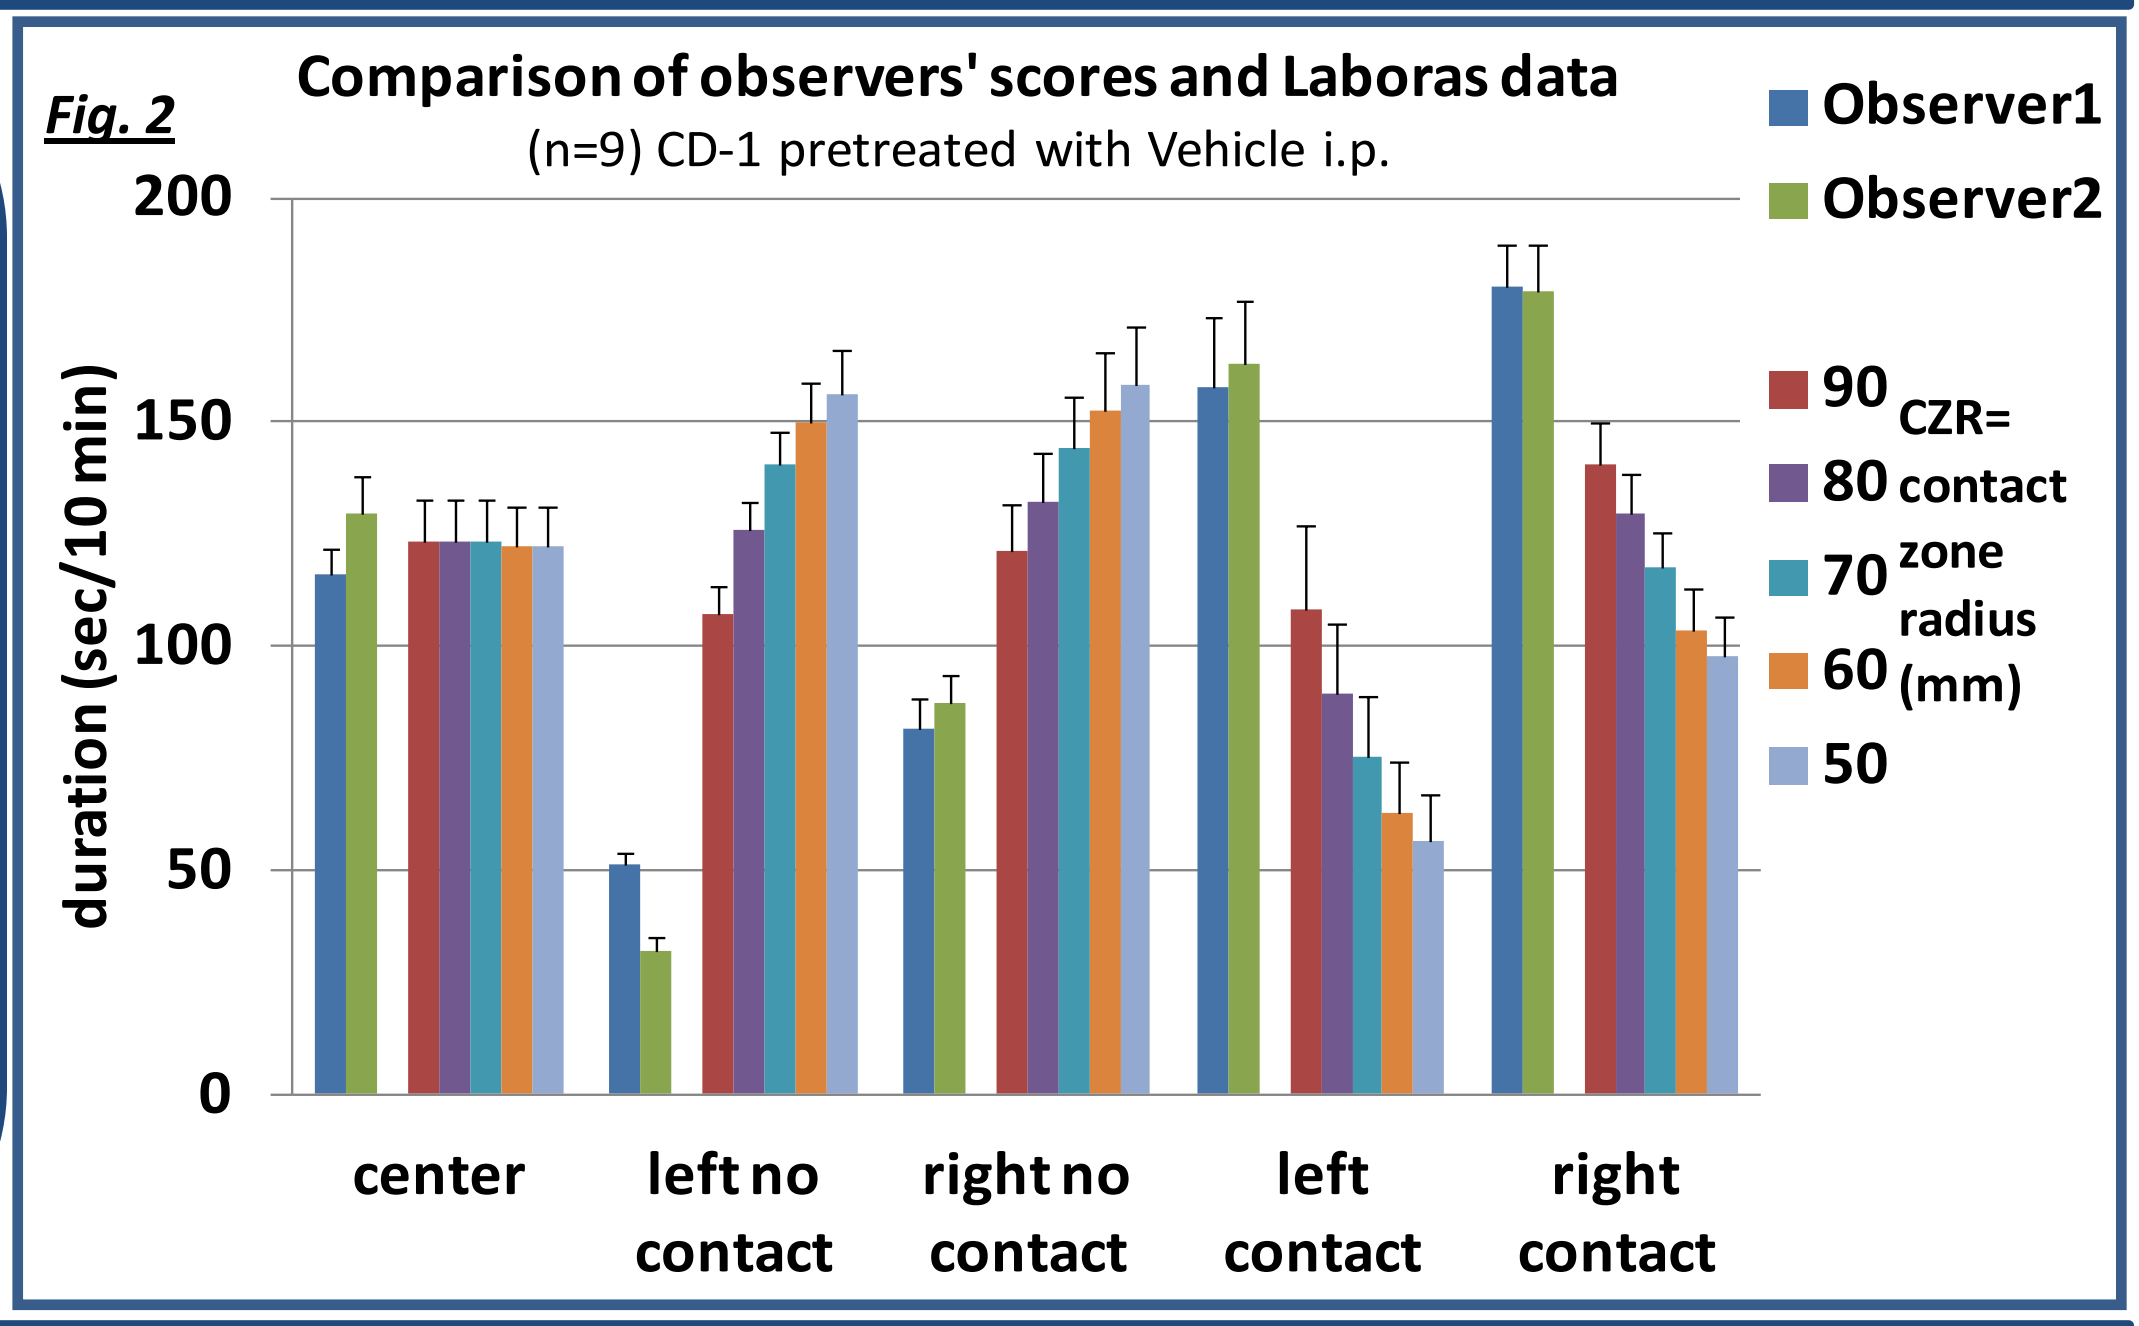

Comparison of observers' scores and Laboras data
(n=9) CD-1 pretreated with Vehicle i.p.
duration (sec/10 min)
200
150
100
50
0
center
left no contact
right no contact
left contact
right contact
Observer1
Observer2
90
80
70
60
50
CZR= contact zone radius (mm)

Conclusion:

1. We validated and optimized a new mouse sociability algorithm of LABORAS system with the comparison of results determined by the system and the scores of two independent blinded observers.
2. We could measure alteration of sociability due to:
 - increase of time spent in contact with social cylinder or
 - decrease of avoidance and less time spent in contact with the empty one.
3. Changes in sociability caused by drugs and at the same time their effect on activity could also be measured with this new sociability set-up in mice.

Measuring sociability in different mouse strains

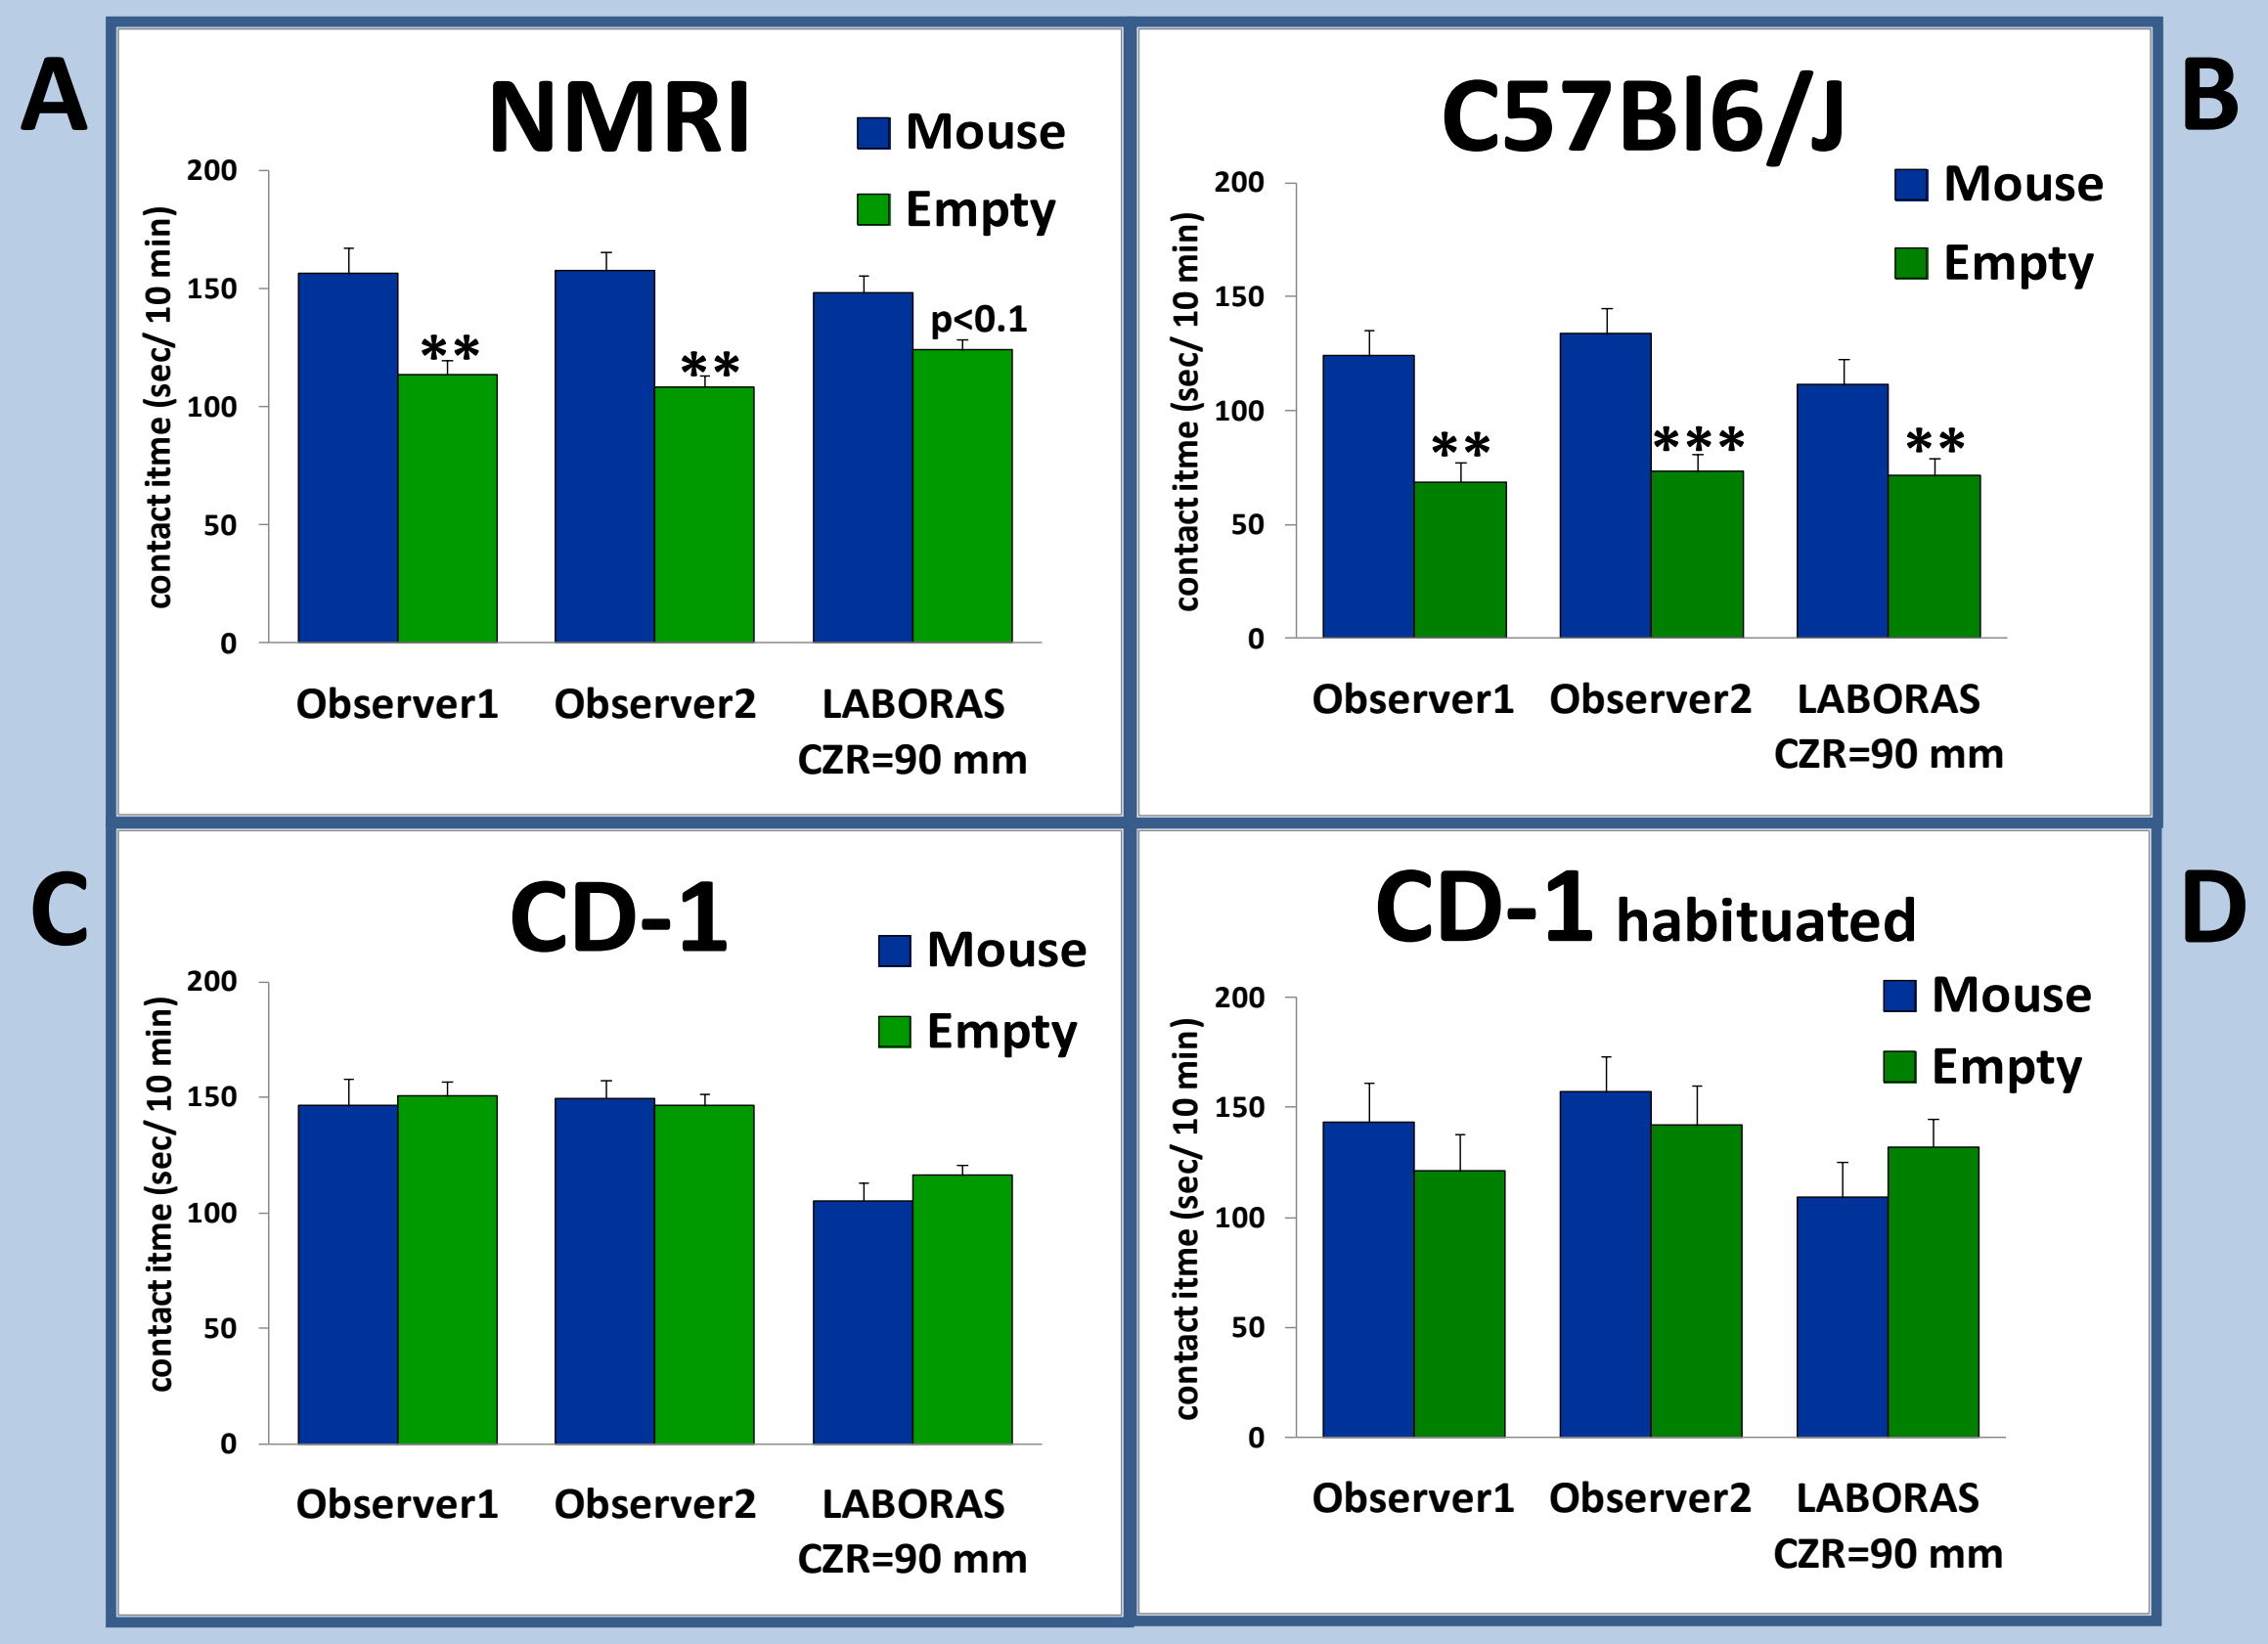

A
NMRI
Mouse
Empty
contact itme (sec/ 10 min)
**
**
p<0.1
Observer1
Observer2
LABORAS CZR=90 mm
B
C57Bl6/J
Mouse
Empty
**

**
Observer1
Observer2
LABORAS CZR=90 mm
C
CD-1
Mouse
Empty
Observer1
Observer2
LABORAS CZR=90 mm
D
CD-1 habituated
Mouse
Empty
Observer1
Observer2
LABORAS CZR=90 mm

Fig.3

Comparison between results of the algorithm and observers' scores.

NMRI (A, n=28) and C57Bl/6J (B, n=16) strains showed normal sociability;
they preferred to spend more time in contact with the social cylinder compared to the empty one.
In contrast to this, CD-1 mice (C, n=44) did not spend more time with the contact of mouse; rather aggressive, often offensive behaviour could be observed. Habituation to the cage (2' in center and 5' in whole) did not modify non-social behaviour of CD-1 mice (D, n=12).

Statistical analysis was performed with paired t-test and with 2-way ANOVA with post hoc Sidak test.
***; p<0.01, ***; p<0.001 vs Mouse contact time*

Aggressive behaviour and sociability

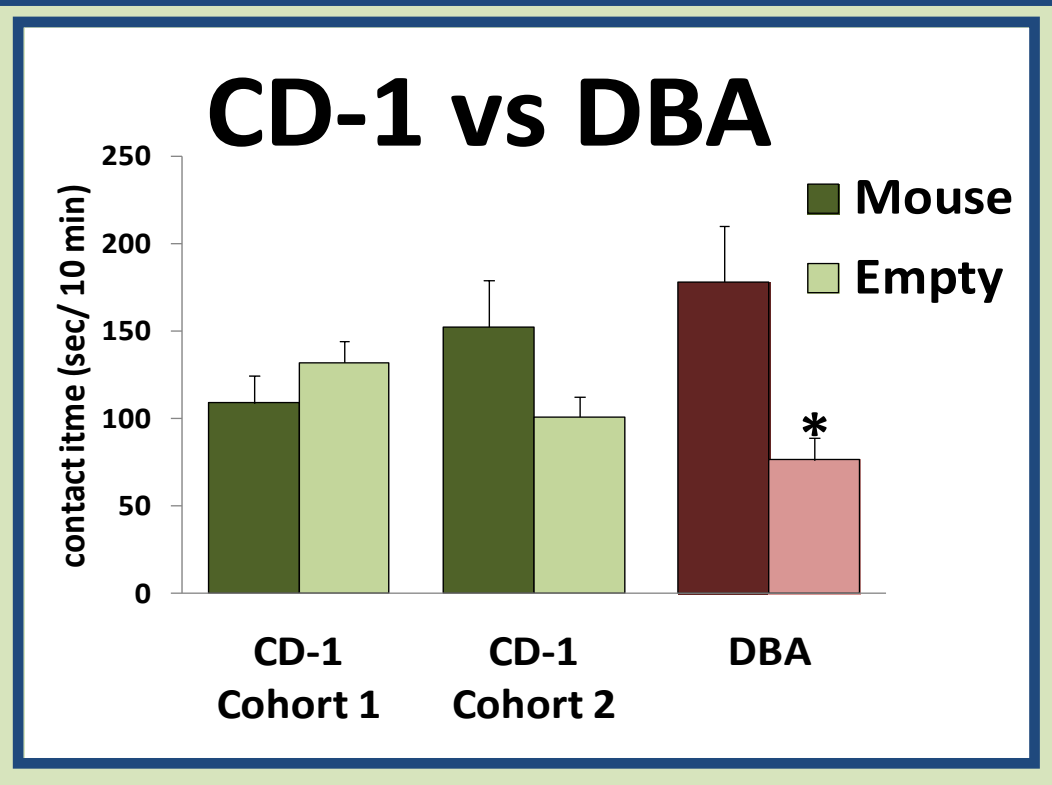

CD-1 vs DBA
Mouse
Empty
contact itme (sec/ 10 min)
250
200
150
100
50
0
*
CD-1 Cohort 1
CD-1 Cohort 2
DBA

Fig.4

CD-1 mice did not prefer social contact and this was not the direct consequence of their offensive nature.

In order to distinguish antisocial and non-social behaviours, an aggressive subgroup of DBA mice was also investigated. Since this subgroup of DBA strain spent more time on the mouse side of chambers, thus the existence of aggressivity alone did not explain avoidance behaviour of the CD-1 strain.

*Mice were 2'+5' habituated. CD-1 Cohort1: n=12, CD-1 Cohort2: n=14, DBA: n=8. Statistical analysis was performed with paired t-test. *; p<0.05 vs Mouse contact time*

Pharmacological manipulation of social behaviour

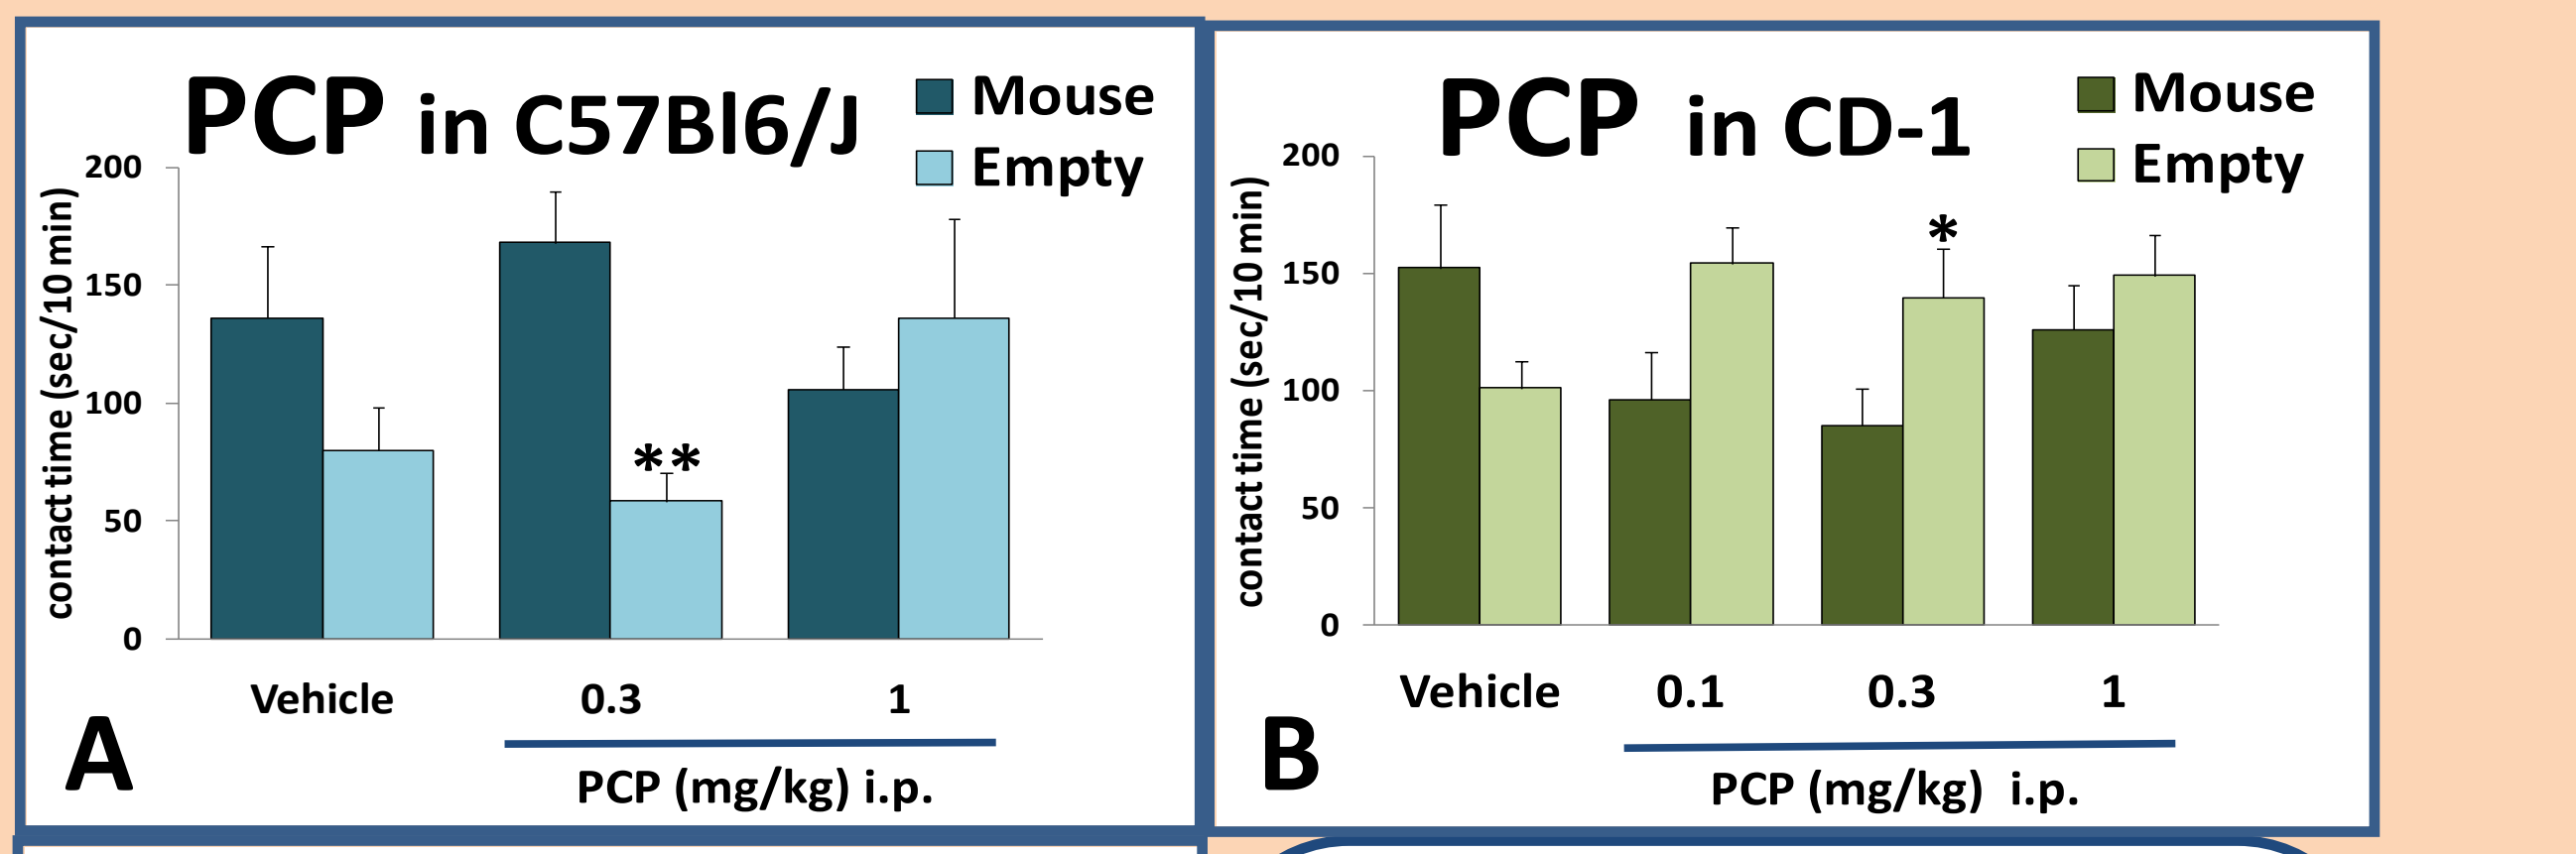

PCP in C57Bl6/J
Mouse
Empty
contact time (sec/10 min)
**
Vehicle
0.3
1
PCP (mg/kg) i.p.
A
PCP in CD-1
Mouse
Empty
*
Vehicle
0.1
0.3
1
PCP (mg/kg) i.p.
B

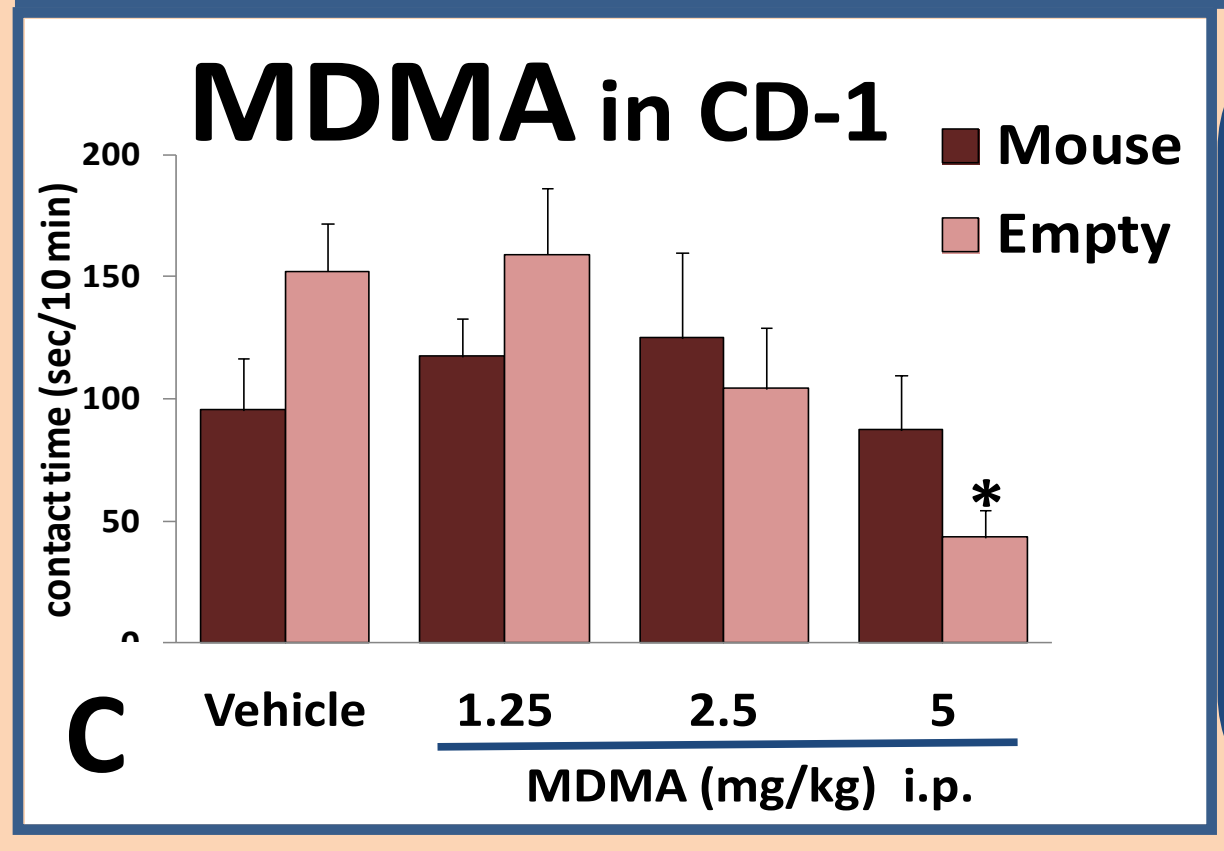

MDMA in CD-1
Mouse
Empty
contact time (sec/10 min)
*
Vehicle
1.25
2.5
5
MDMA (mg/kg) i.p.
C

	Dose (mg/kg) i.p.	n=	Distance travelled (m) mean	SEM	change%
PCP in C57Bl6/J	Vehicle	11	5.72	0.40	-
	0.3	7	6.94	0.80	24
	1	10	6.89	0.88	23
PCP in CD-1	Vehicle	14	10.24	1.12	-
	0.1	12	10.93	0.67	7
	0.3	12	9.22	1.10	-10
	1	12	11.05	0.67	8
MDMA in CD-1	Vehicle	12	8.76	0.66	-
	1.25	10	10.85	0.76	24
	2.5	11	10.41	0.99	19
	5	12	9.75	1.13	11

Fig. 5.

PCP deteriorated whereas MDMA ameliorated decreased social behaviour of mice.

In C57Bl6/J mice PCP at the dose of 0.3 mg/kg did not modify social preference, while 1 mg/kg dose impaired it and produced similar contact time on both sides (A). In CD-1 mice PCP (0.1-1 mg/kg) also showed this social impairment effect started at lower dose and it seemed to increase their avoidance behaviour. At the dose of 1 mg/kg the initiation of activity increasing mechanism might cause the elevation of contact time on both sides (B).

MDMA, similarly to its prosocial effect observed in neurotypical humans [4] was able to ameliorate decreased social behaviour of CD-1 mice in a dose-dependent manner via decrease of contact time with the empty cylinder (C).

*Mice were 2'+5' min habituated. Statistical analysis was performed with paired t-test or 2-way ANOVA with post hoc Sidak test. *; p<0.05, **; p<0.01 vs Mouse contact time*

References:

[1] Moy, S.S., Nadler, J.J., Perez, A., Barbaro, R.P., Johns, J.M., Magnuson, T.R., Piven, J., Crawley, J.N., 2004. Sociability and preference for social novelty in five inbred strains: an approach to assess autistic-like behavior in mice. Genes Brain Behav 5, 287-302.

[2] Moy, S.S., Nadler, J.J., Young, N.B., Perez, A., Holloway, L.P., Barbaro, R.P., Barbaro, J.R., Wilson, L.M., Threadgill, D.W., Lauder, J.M., Magnuson, T.R., Crawley, J.N., 2007. Mouse behavioral tasks relevant to autism: phenotypes of 10 inbred strains. Behav Brain Res176, 4-20.

[3] Quinn, L.P., Stean, T.O., Trail, B., Duxon, M.S., Stratton, S.C., Billinton, A., Upton, N., 2003. LABORAS: Initial pharmacological validation of a system allowing continuous monitoring of laboratory rodent behaviour. J Neurosci Methods 130, 83-92.

[4] Danforth, A.L., Struble, C.M., Yazar-Klosinski, B., Grob, C.S., 2016. MDMA-assisted therapy: A new treatment model for social anxiety in autistic adults. Prog Neuropsychopharmacol Biol Psychiatry 64, 237-249.

This work was supported by a Hungarian governmental grant (ERNYO-13-1-2013-0003).

Authors of Gedeon Richter have no potential conflict of interest (grant, honorary, advisory board etc.) from Metris b.v.

Copyright © 2016 Krisztina Sz. Kordás, Viktor Román; Laboratory of Neurodevelopmental Biology, Gedeon Richter Plc., 19-21 Gyömrői street, 1103-Budapest, Hungary; Herman Eldering, Ronald Bulthuis; Metris b.v., Kruisweg 825, 2132 NG Hoofddorp, the Netherlands